Polyhydroxy acid-based topical treatment for ichthyosis in the golden retriever: histopathological findings

A. Puigdemont*, N. Furiani†, D. Fondevila*, L. Ramió-Lluch§, and P. Brazís§

Universitat Autònoma de Barcelona, Bellaterra, Barcelona, Spain.
†Studio di Dermatologia Veterinaria, Perugia, Italy.
§Laboratorios LETI, Barcelona, Spain.

Introduction

Autosomic recessive congenital ichthyosis (ARCI) in golden retrievers has been associated with a mutated PNPLA1 gene.
ARCI typically manifests as generalized scaling of skin giving rise to scales of different size and shades of white or black, mainly in the ventral and lateral region of the neck and trunk[1].

Microscopically, the main feature of ARCI is laminar orthokeratotic hyperkeratosis with minimal to no epidermal hyperplasia and the absence of an inflammatory reaction. The presence of scattered keratinocytes containing clear intracellular spaces (vacuoles) in the outer spinous layer is also considered characteristic.[1,2,3]

Among other polyhydroxy acid, gluconolactone promotes *stratum corneum* acidification improving the epidermal barrier through epidermal lipid maturation and keratinocyte cohesion. [4,5]

This study examines the efficacy of a gluconolactone and other α and ß hydroxy acids topical treatment for the management of ichthyosis in golden retrievers.

Material and methods

Two female golden retrievers of 6 years old with clinical signs of ARCI and a PCR-confirmed mutated PNPLA1 gene, received a shampoo and a lotion containing gluconolactone and other α and ß hydroxy acids (Kerato, Laboratorios LETI, Spain) twice a week for 15 days and then weekly for other 15 days. Biopsy specimens were obtained from the ventro-lateral aspect of the thorax before and 30 days after starting treatment. Reductions in the extent and size of the scales were assessed by the veterinary dermatologist on days 15 and 30 in comparison to day 0. Improvements were expressed as percentage.

Results

Before treatment, histological findings revealed a compact orthokeratotic hyperkeratosis and multifocal pigmentation. On day 30 orthokeratotic hyperkeratosis was reduced, keratin was re-organized as a laminated basket weave mesh and pigmentation decreased. However, as expected, vacuolated cells were still present in the examined samples (Fig. 1).

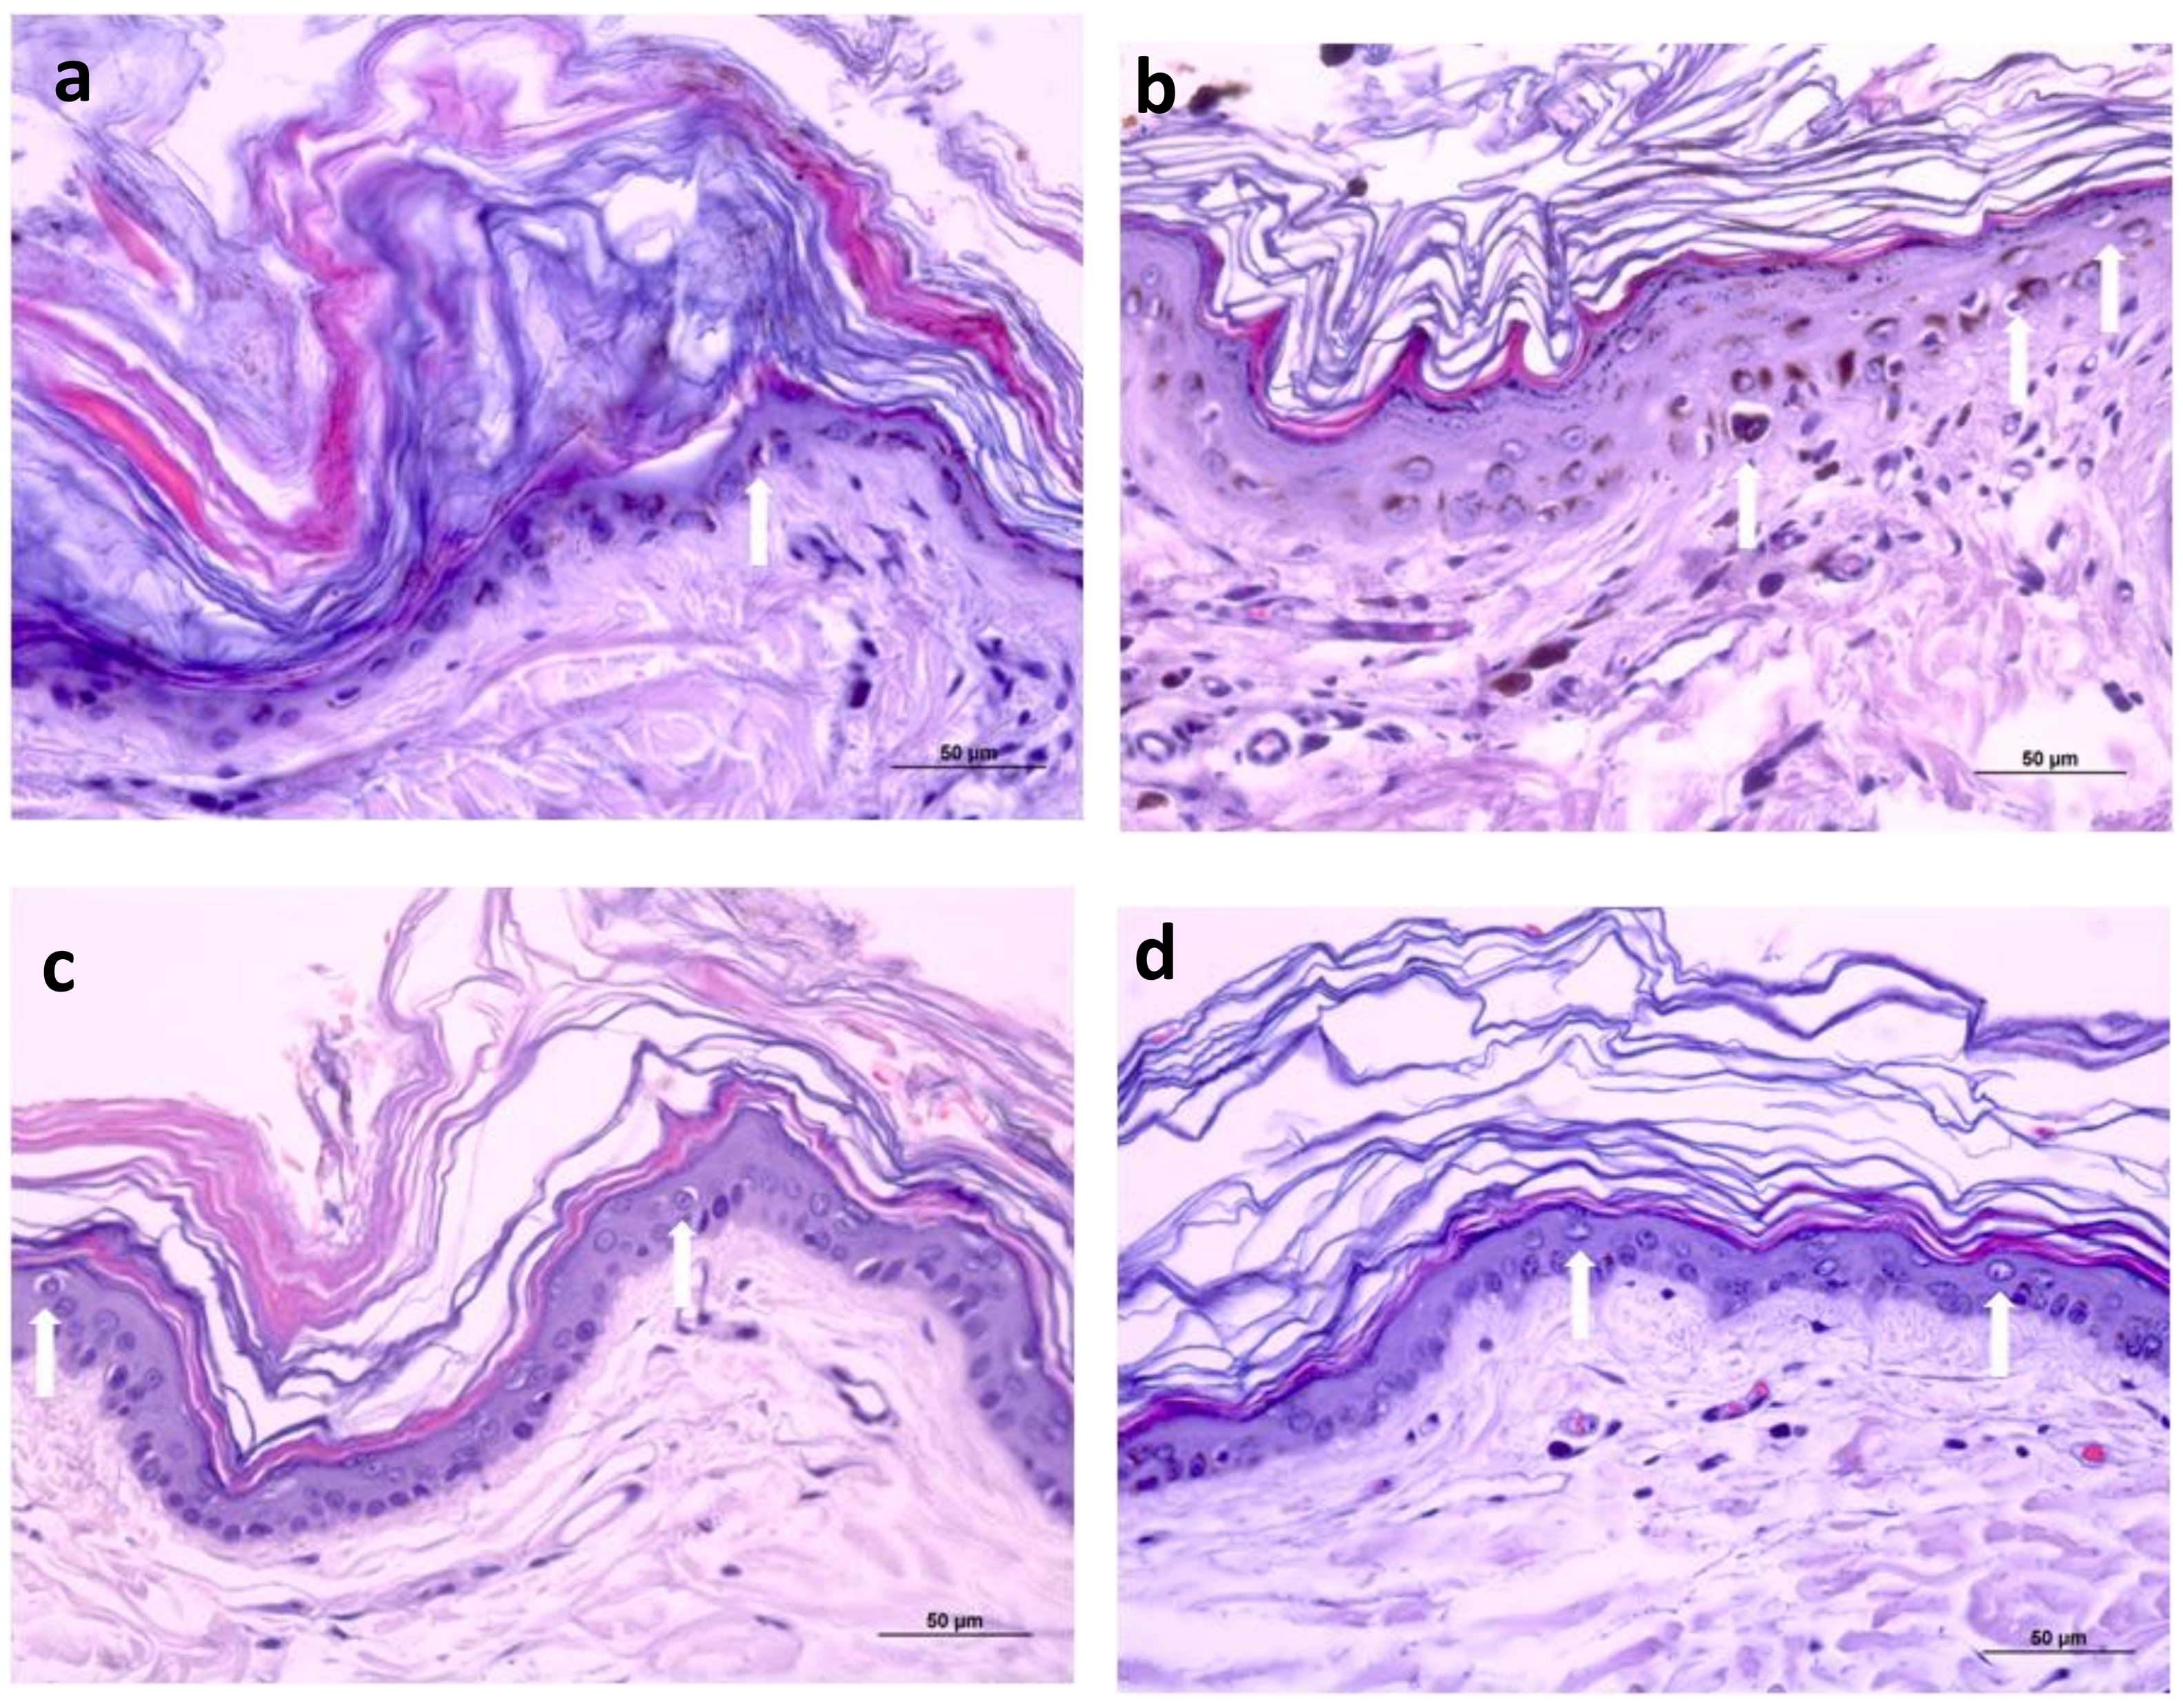
a
b
c
d
50 µm
50 µm
50 µm
50 µm

Figure 1. Skin biopsies before treatment (a, c) with diffuse laminar orthokeratotic hyperkeratosis in the *stratum corneum* and scattered vacuolated keratinocytes in the *stratum granulosum* and *spinosum*. After 30 days of topical treatment, keratin recovers its typical basket weave appearance (b, d). Haematoxylin and eosin.

At days 15 and 30, the topical treatment had significantly reduced the presence of the scales by 60% and 90%, respectively (Fig. 2).

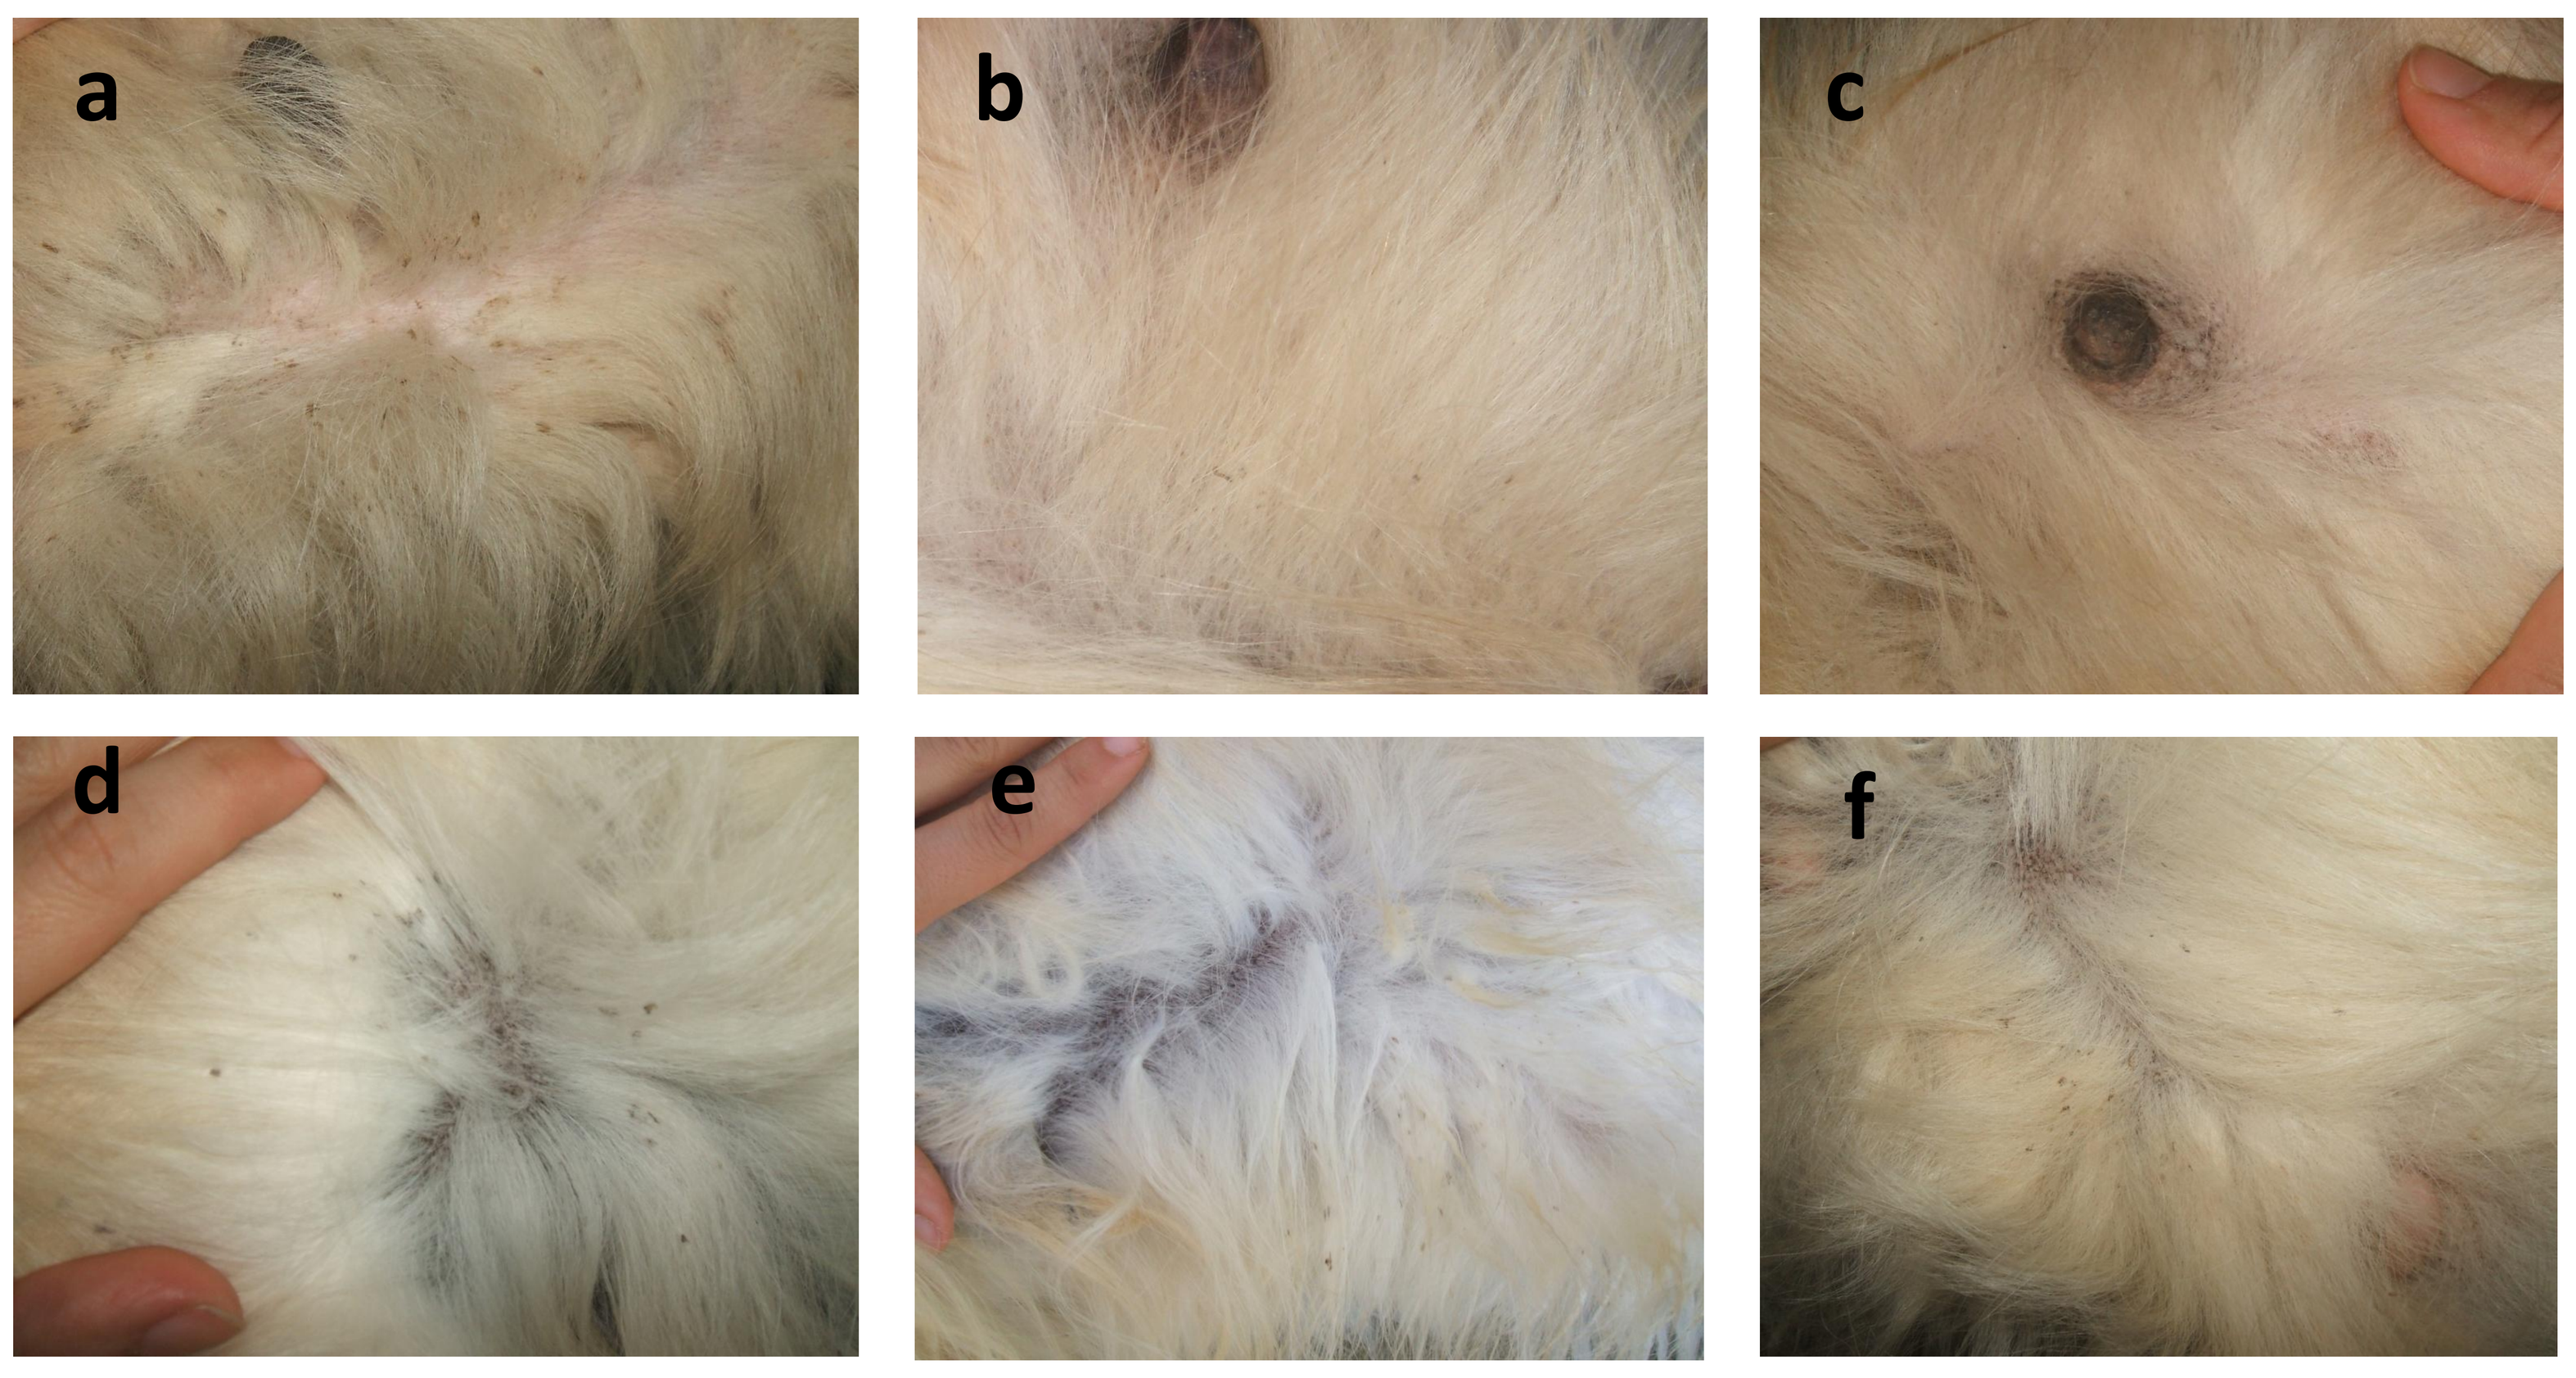
a
b
c
d
e
f

Figure 2. Clinical response . Pictures obtained before (a, d), after 15 days (b,e) and 1 month of treatment (c, f)).

Conclusion

A topical treatment with gluconolactone and α, ß hydroxy acids improved the condition of the *stratum corneum* and reduced the number of scales in golden retrievers with ARCI after 30 days of treatment.

Bibliography:

1- Guaguere E, et al. Clinical, histopathological and genetic data of ichthyosis in the golden retriever: a prospective study. *Journal of Small Animal Practice* 2009, 50:227–235.
2- Pin D, et al.. An emulsion restores the skin barrier by decreasing the skin pH and inflammation in a canine experimental model. J Comp Pathol 2014, 151:244-54.
3-Tamamoto-Mochizuki C et al. Autosomal recessive congenital ichthyosis due to PNPLA1 mutation in a golden retriever-poodle crossbred dog and the effect of topical therapy. Vet Dermatol 2016, 27:306-e75
4-Berardesca E, et al. Alpha hydroxyacids modulate stratum corneum barrier function. *Br J Dermatol* 1997, 137:934-938
5-Hachem JP et al. Acute acidification of stratum corneum membrane domains using polyhydroxyl acids improves lipid processing and inhibits degradation of corneodesmosomes. J Invest Dermatol 2010, 130:500-10.