Characterization of the allergenic profile from a *D. farinae* extract used in immunotherapy for dogs.

R. Moya[1], J. Carnes[1], N. Sinovas[1], L. Ramió-Lluch[2], P. Brazis[2], A. Puigdemont*[3]

[1]R&D Department, Laboratorios Leti. Barcelona , Spain.
[2]UNIVET, Barcelona, Spain.
[3]Department of Pharmacology, Therapeutics and Toxicology, Universitat Autònoma de Barcelona, Bellaterra., Spain

Introduction

House dust mites have been identified as the main non-seasonal agents responsible for canine atopic dermatitis (CAD)[1]. Unlike human allergic patients, the allergens of groups 1 and 2 have been described as minor allergens for dogs, whereas allergens of groups 15 and 18 are considered the major allergens [2-4].

Objectives

- To characterise a commercial *D. farinae* extract, in order to evaluate whether the extracts contain Der f 15 and Der f 18.
- To investigate the allergenic profile of sensitization and the immunological characteristics of a population of dogs suffering from atopic dermatitis

Material and methods

Eighteen atopic and 3 healthy dogs were included in the study. Atopic dogs were diagnosed by the accepted clinical criteria[5] and presented positive specific IgE against *D. farinae* (>2500 ELISA A.U; Greer Laboratories, Lenoir, NC; USA). Clinical characteristic of the population are represented in Table 1.

D. farinae allergen extract was deeply characterized by SDS-PAGE and canine major allergens, Der f 15 and Der f 18, as well as Der f 1 and Der f 2 were identified by mass spectrometry and quantified by scanning densitometry.

The allergenic profile of each dog was investigated by immunoblot using *D. farinae* extract in solid phase (50 µg/serum sample). The specific IgE and IgG subclasses were measured by direct ELISA. A cut-off of 0.35 OD (mean plus 2 standard deviations of control dogs values) was established for specific IgE, while values ≥ 0.1 were considered positive for specific IgG, IgG1 and IgG2.

Table 1. Characteristics of the dog population included in the study.

Number	IgE D. farinae (EAU)	Breed	Age (years old)	Sex
1	2526	Shar Pei	4	Female
2	2531	Crossbred	5	Male
3	3347	Staffordshire Bull Terrier	5	Male
4	3157	Yorkshire Terrier	7	Female
5	3136	German Shepherd	3	Female
6	2551	Beagle	3	Female
7	3408	Shih-Tzu	6	Male
8	3857	Fos Terrier	5	Female
9	2627	Pit Bull Terrier	6	Female
10	2946	West Highland White Terrier	10	Female
11	2807	American Pitbull	1	Male
12	3247	Perro de aguas- spaniel	5	Female
13	3423	Labrador	6	Male
14	2564	Labrador	6	Male
15	3403	Labrador	7	Male
16	2959	Catalan Shepherd	4	Female
17	2520	Argentine Dogo	1	Male
18	2910	Crossbred	2	Female
C1	39	Chinese crested	1	Male
C2	30	Shiba-Inu	2	Female
C3	0	Beagle	2	Female

Results

Der f 15 and Der f 18 as well as Der f 1 and Der f 2 were identified in whole extract (Figure 1A). Additionally, identity of Der f 15 and Der f 18 was confirmed by mass-spectrometry. The protein content of these allergens in the extract was determined and is showed in Figure 1B.

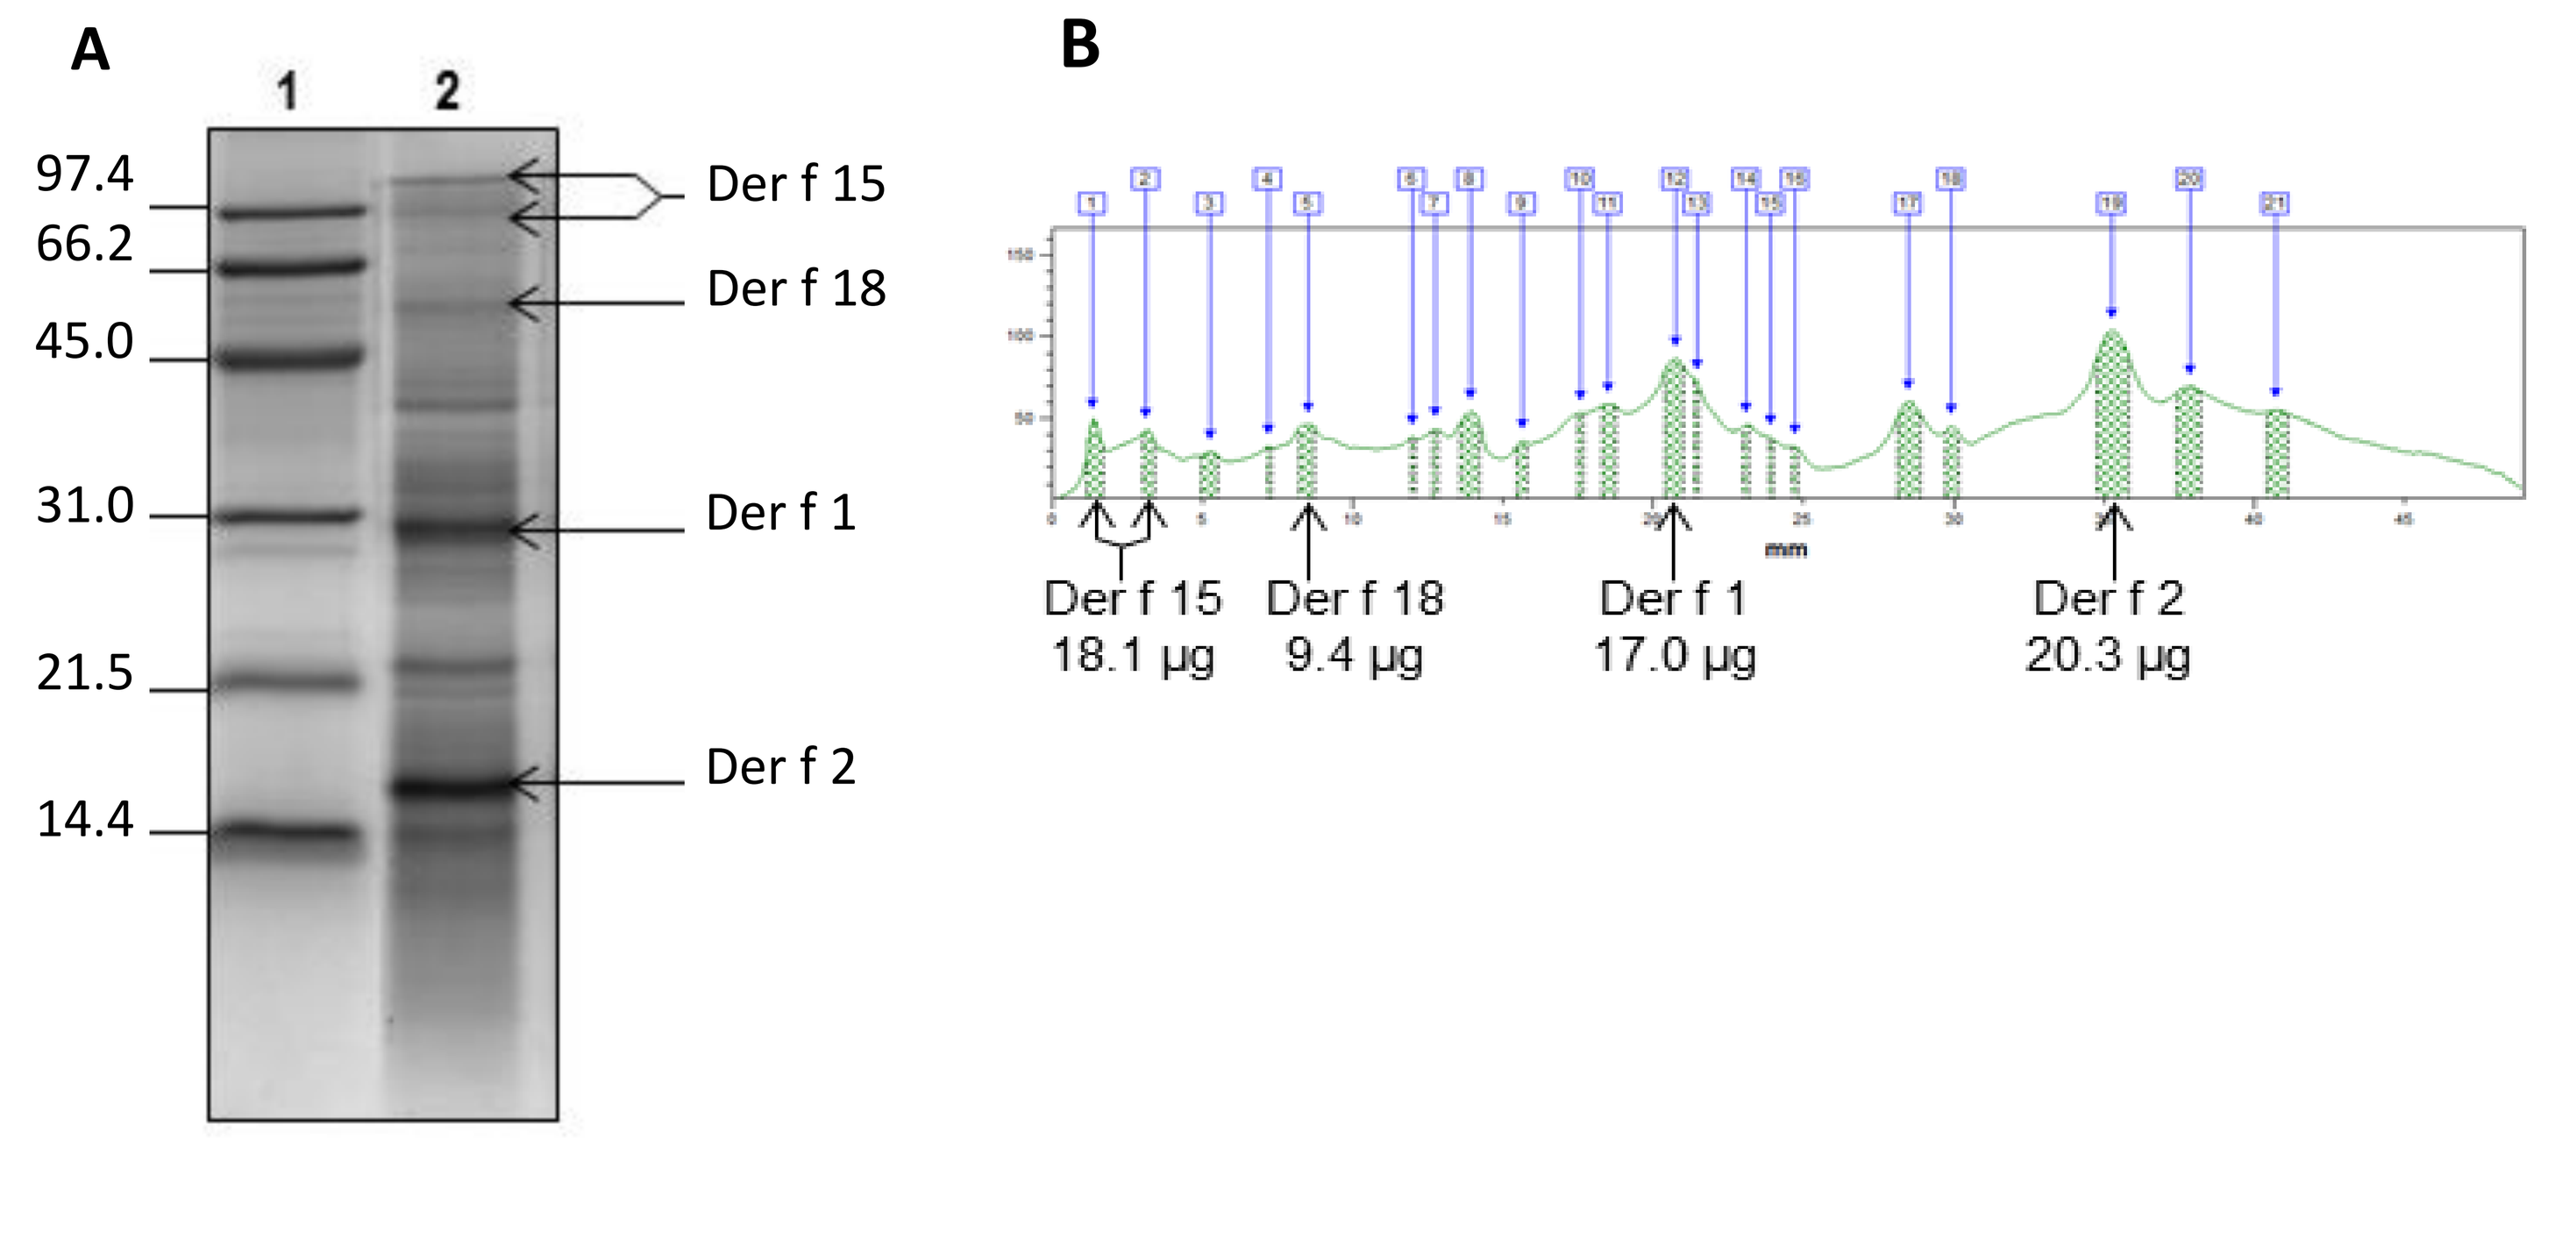
A
1
2
97.4
66.2
45.0
31.0
21.5
14.4
Der f 15
Der f 18
Der f 1
Der f 2
B
Der f 15 18.1 µg
Der f 18 9.4 µg
Der f 1 17.0 µg
Der f 2 20.3 µg

Figure 1: SDS-PAGE stained with Coomasie gels (A). 1: MW marker; 2: D. farinae extract (8 µg of protein). B) Band analysis of gel with ImageQuant software; µg of allergen/mg of lyophilized extract of Der f 15, Der f 18, Der f 1 and Der f 2 are indicated below their corresponding band.

As is observed in Figure 2, immunoblot assays confirmed the recognition of allergens of groups 15 and 18 but also from groups 1, 2 and other medium molecular weight allergens in CAD.

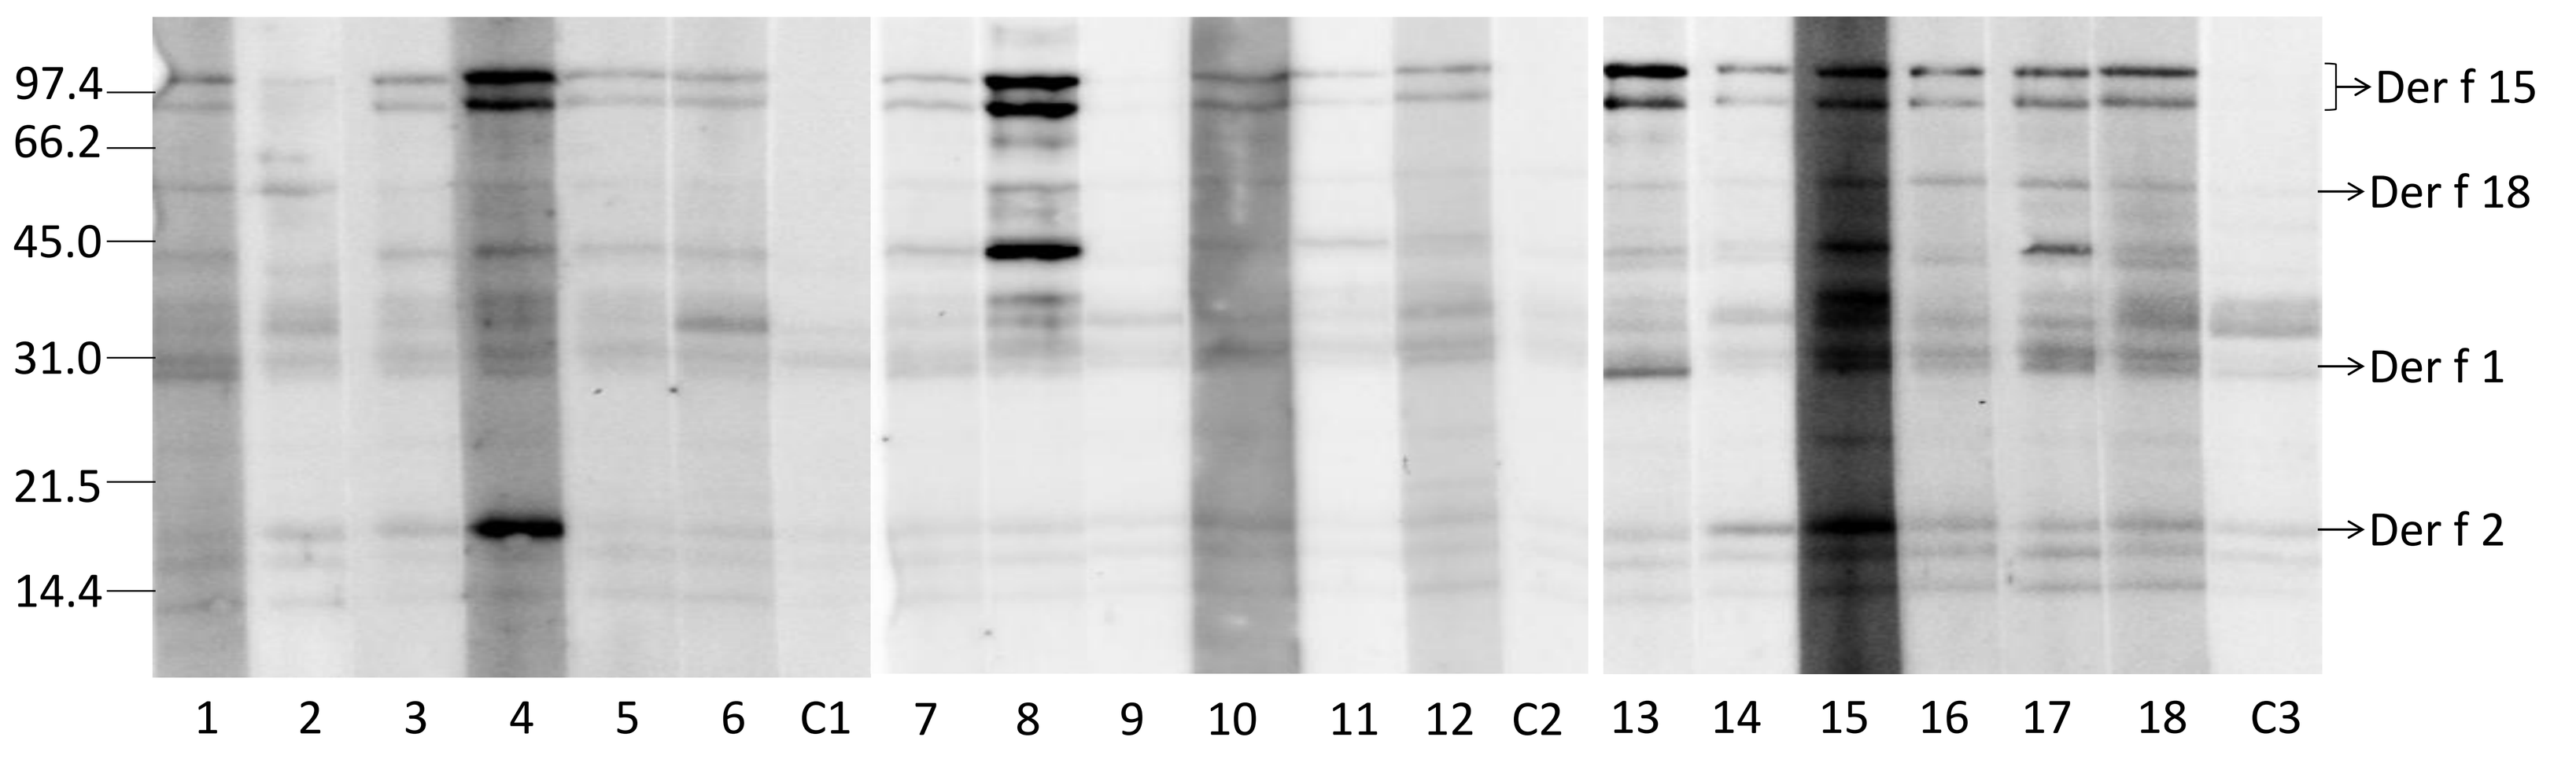
97.4
66.2
45.0
31.0
21.5
14.4
Der f 15
Der f 18
Der f 1
Der f 2
1 2 3 4 5 6 C1 7 8 9 10 11 12 C2 13 14 15 16 17 18 C3

Figure 2: Immunoblot of 50 µg of *D. farinae* extract run and incubated with positive sera (1-18) and control (C1, C2 and C3). Molecular weight marker is indicated on the left.

The 18 sensitized dogs showed a positive specific IgE against *D. farinae extract,* being the OD in the range of 0.4-3.6. The 3 negative controls showed negative absorbance values, below 0.25 (Figure 3). Concerning specific IgG and IgG subclasses, the IgG1 was the only immunoglobulin where a difference was observed between the control and the sensitized group. Absorbance values obtained for specific IgG and IgG2 were in general higher in sera from sensitized dogs, although the negative controls sera were also found to be positive for these immunoglobulins (Figure3).

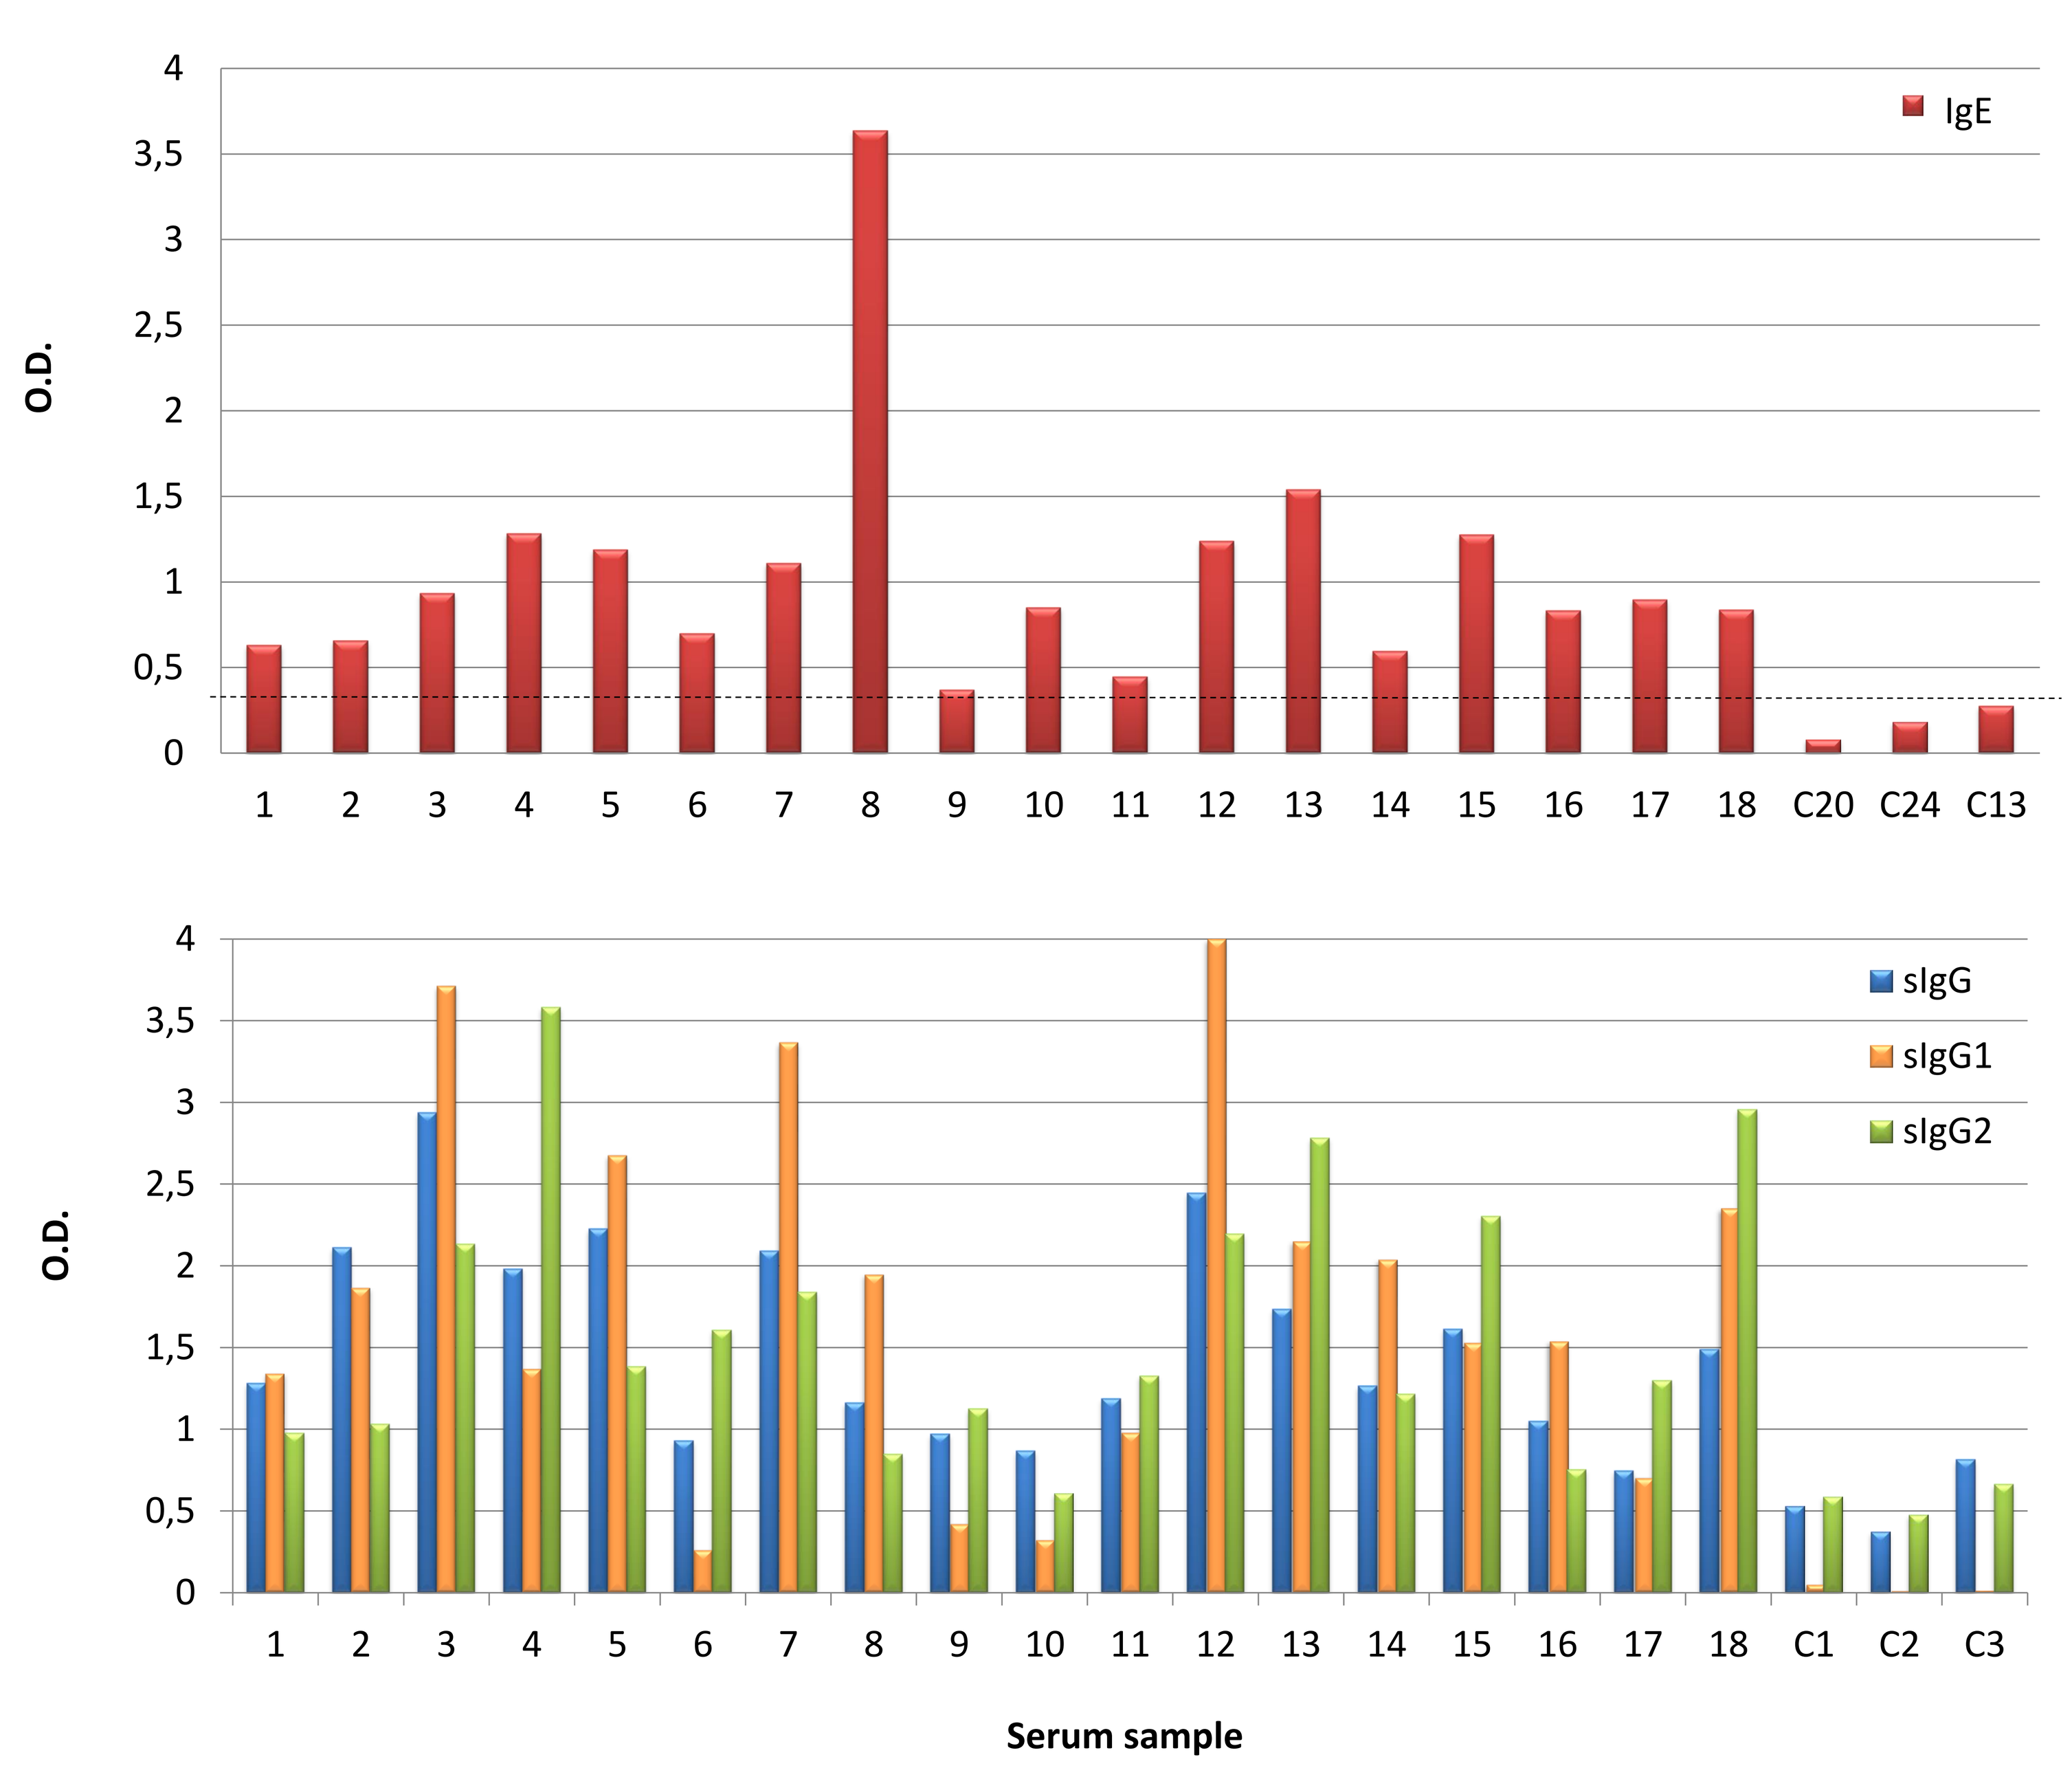
A
O.D.
IgE
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 C20 C24 C13
O.D.
sIgG
sIgG1
sIgG2
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 C1 C2 C3
Serum sample

Figure 3: Specific immunoglobulins levels measured by ELISA in serum samples of atopic (1-18) and control dogs (C1-3) using 2 µg of *D. farinae extract in the solid phase. A) Specific* IgE (serum samples diluted 1:8). Cut-off is marked with a dotted line. B) Specific IgG, IgG1 and IgG2 (serum samples diluted 1:2000).

Conclusions

Allergenic profile of dogs show a sensitization to major allergens Der f 15 and Der f 18 but also to Der f 1 and Der f 2. Therefore, the extract used in the present study is an optimal candidate for both diagnosis and immunotherapy of mite allergens in dogs.

References

1. Nuttall TJ, Hill PB, Bensignor E et al. 2006. Vet Dermatol 17, 223-35.
2. Noli C, Bernardina WE, Willemse T. 1996. Vet Immunol Immunopathol 52, 147-57.
3. Goicoa A, Espino L, Rodriguez I et al. 2008. Acta Vet Hung 56, 163-71.
4. Hou CC, Griffin CE, Hill PB. 2008. Vet Dermatol 19, 215-20.
5. Favrot C, Steffan J, Seewald W et al. 2010. Veterinary Dermatology 21, 23–30.

LETI

Universitat Autònoma de Barcelona